AnelloVector™ Viral Vectors: encapsidating anellovirus-based vectors beyond the wild type genome capacity and demonstrating 12-month durable expression of an eGFP transgene post subretinal administration

Cato Prince, George Bounoutas, Bolu Zhou, Waseem Raja, Isabella Gold, Rianna Pozsgai, Parmi Thakker, Nicole Boisvert, Christopher Reardon, Stephanie Thurmond, Erin Ozturk, Emily Stead, Rajendra Boggavarapu, Simeon Springer, Lovepreet Chahal, Maciej Nogalski, Tuyen Ong, Christopher Wright, Ashley Mackey, Geoffrey Parsons, Joseph Cabral
Ring Therapeutics, Cambridge, MA, USA

Abstract

Anelloviridae is a family of non-enveloped viruses with negative-sense, circular, single-stranded deoxyribonucleic acid (ssDNA) genomes that infect vertebrates and are a ubiquitous component of the human virome. Human anelloviruses evade induction of humoral immune responses and appear to be non-pathogenic. These properties, in conjunction with their enormous genomic diversity and wide tissue distribution, make anelloviruses compelling candidates as vectors for next-generation genetic medicines. Here we report the first gene delivery vector system based on a human commensal virus: the *AnelloVector™* viral vector. The *AnelloVector™* viral vector is based on a virus of the *Betatorquevirus* genus. Production is enabled by the development of the Self-Amplifying Trans-complementation of a Universal Recombinant aNellovector™ (SATURN) system, which relies on a self-replicating plasmid to provide viral proteins in trans that drive replication and capsid-dependent packaging of vector genomes. The SATURN system also utilizes a Cre-lox-based recombination mechanism to generate single unit-sized circular genomes inside the MOLT-4 production cell line. Anelloviruses are the only truly negative-sense ssDNA viruses that infect eukaryotes. Notably, anelloviruses lack the Rep proteins produced by other ssDNA viruses, pointing to a unique replication and packaging mechanism that may allow for the encapsidation of larger genomes. The *AnelloVector™* viral vector presented here is based on the Beta genera of human anelloviruses which have genome sizes of ~2.8-3.0 kb. Using vector genomes of increasing size for encapsidation, we have demonstrated that *AnelloVector™* viral vectors are capable of packaging vector genomes ranging from 2.0kb - 5.0kb, representing up to 65% beyond the wild-type genome capacity. The *AnelloVector™* viral vector demonstrated function *in vitro* in retinal pigment epithelial (RPE) cells, the cell type from which this betatorquevirus was originally discovered. We next investigated the potential of the *AnelloVector™* viral vector to serve as platform capable of durable gene expression. Using an eGFP transgene to track expression, the *AnelloVector™* viral vector demonstrated durable *in vivo* expression in the mouse eye for 12 months following subretinal administration. *AnelloVector™* viral vectors have great potential to deliver safe, durable, redosable, and potent therapeutics, helping to expand the reach of programmable medicines.

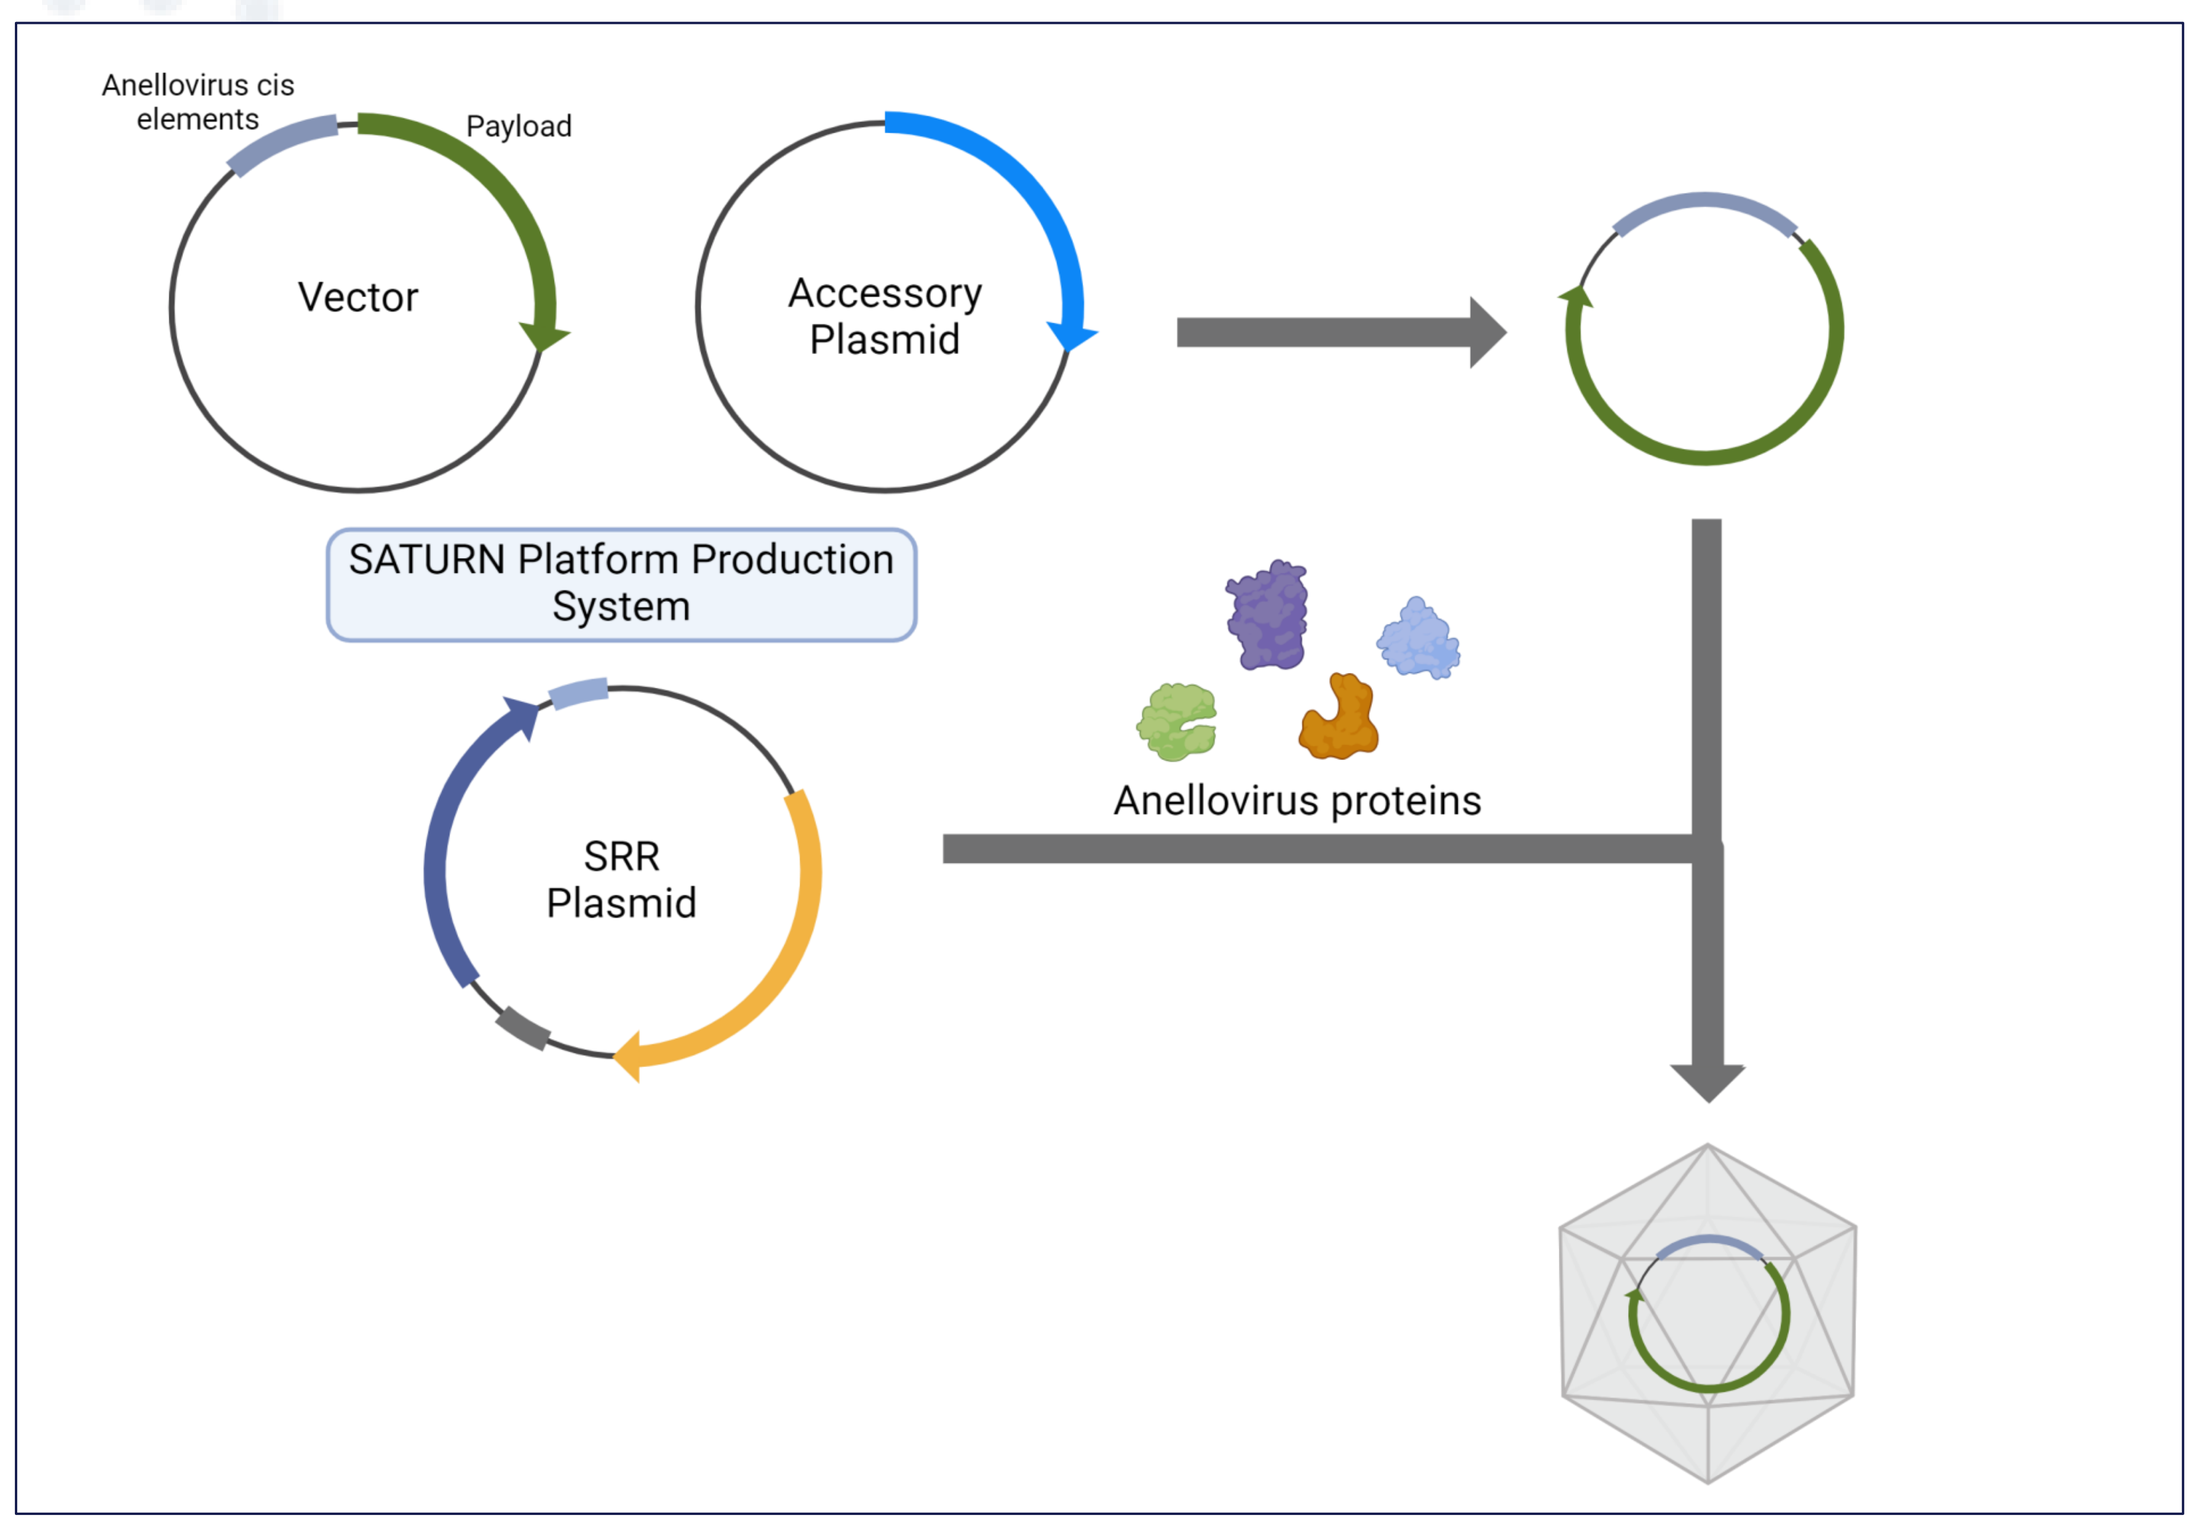
Anellovirus cis elements
Payload
Vector
Accessory Plasmid
SATURN Platform Production System
SRR Plasmid
Anellovirus proteins

Figure 1: Representation of *AnelloVector™* viral vector production using a three-plasmid transfection system. The triple transfection comprises the vector plasmid containing anellovirus cis elements and transgene flanked by lox sites, a Cre expression plasmid (accessory plasmid), and a self-replicating rescue plasmid (SRR plasmid) which provides anellovirus proteins in trans.

Proposed model for ANV packaging

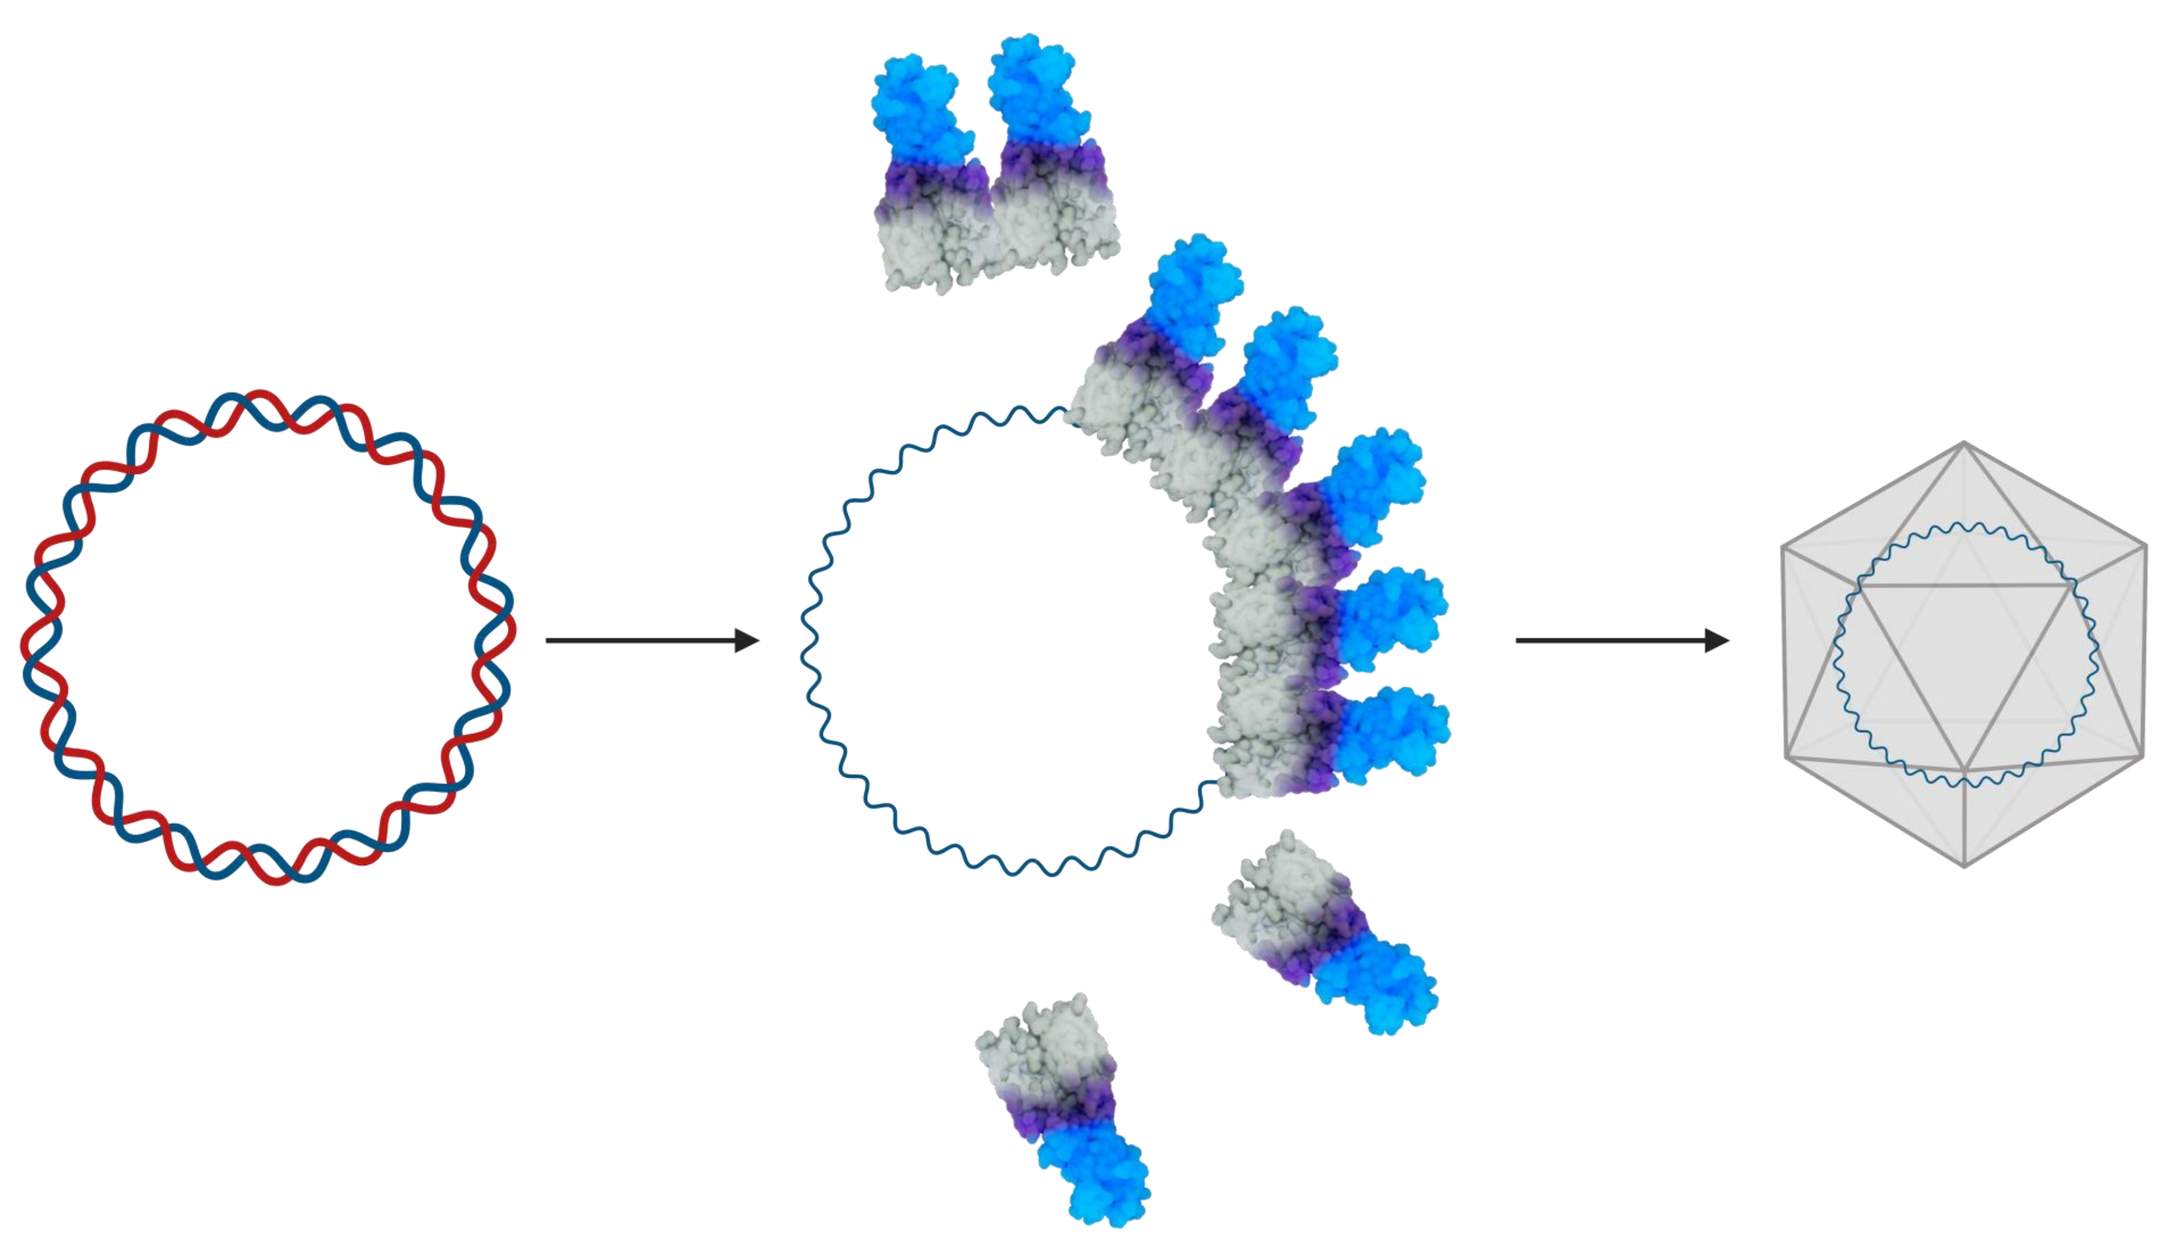

Figure 2: Diagram representing the proposed model for Anellovirus particle formation. The double-stranded intermediate of the anellovirus genome expresses nonstructural and capsid proteins. Capsid proteins begin to spontaneously assemble around replicated or actively-replicating cssDNA. Virions are formed containing cssDNA anellovirus genome and 60 subunits of the capsid protein.

AnelloVector™ viral vectors can encapsidate larger than wild type payloads - up to 5.0kb

A

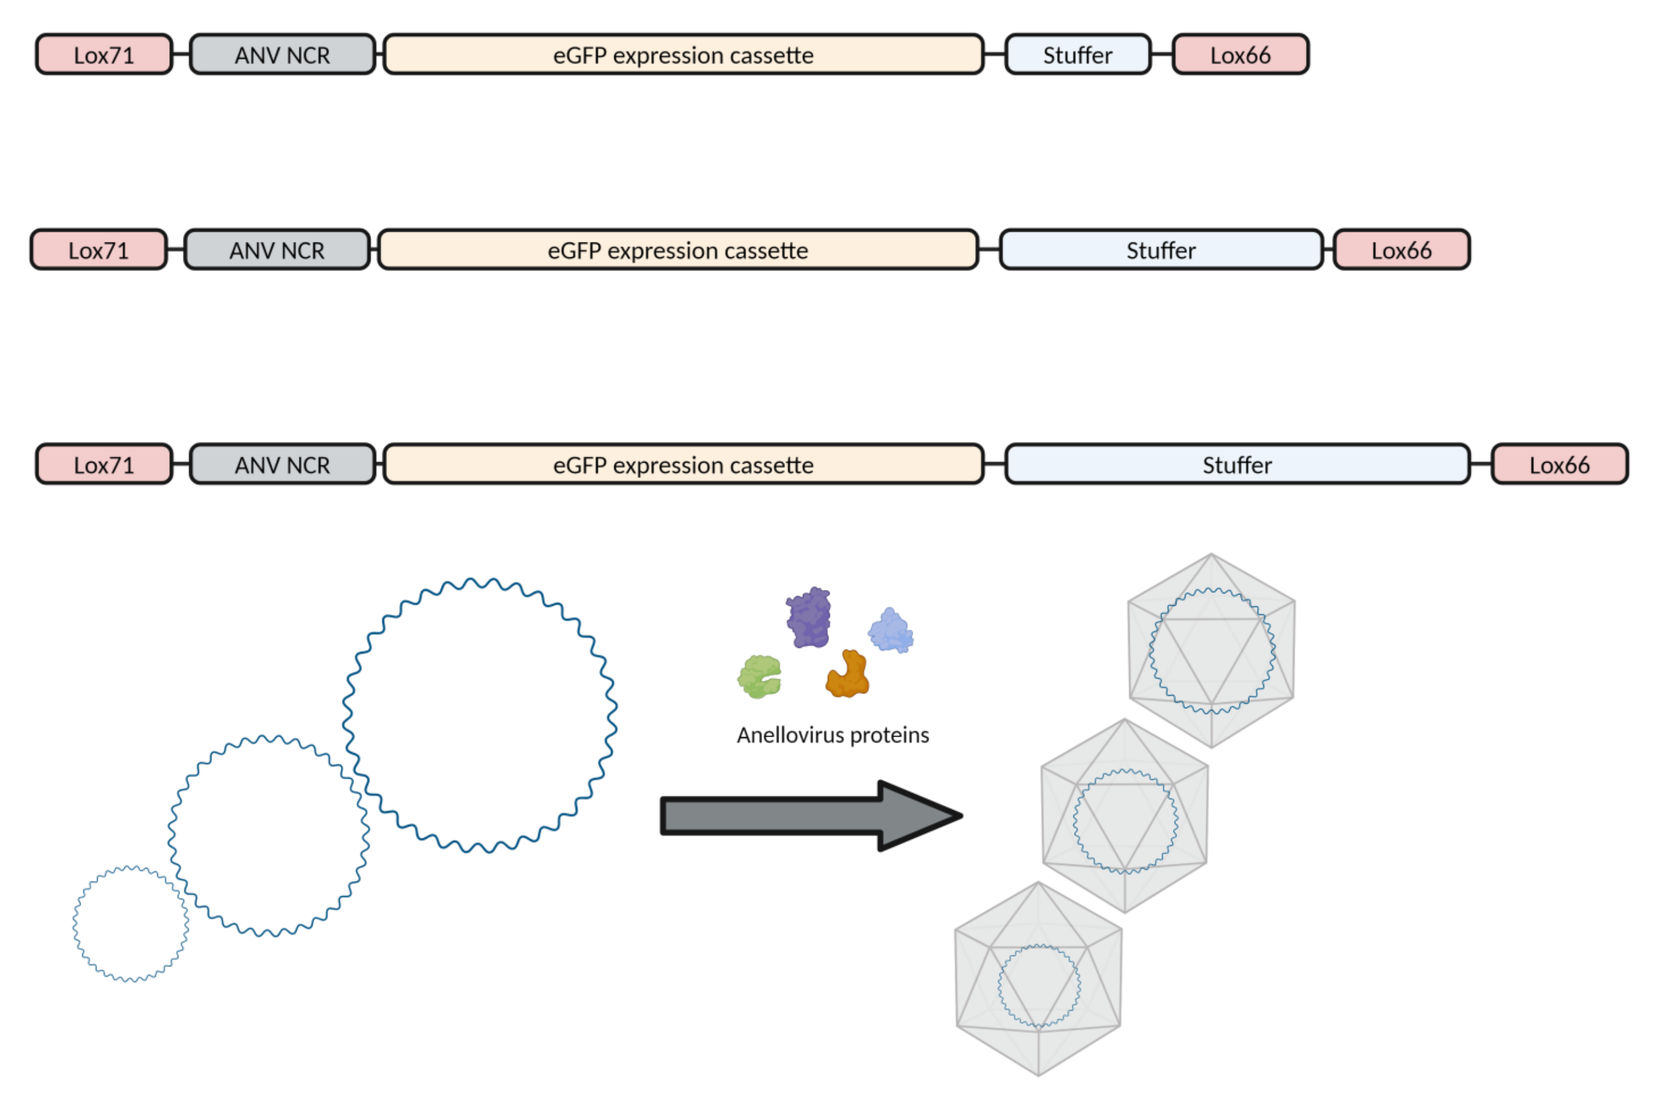
Lox71
ANV NCR
eGFP expression cassette
Stuffer
Lox66
Anellovirus proteins

B

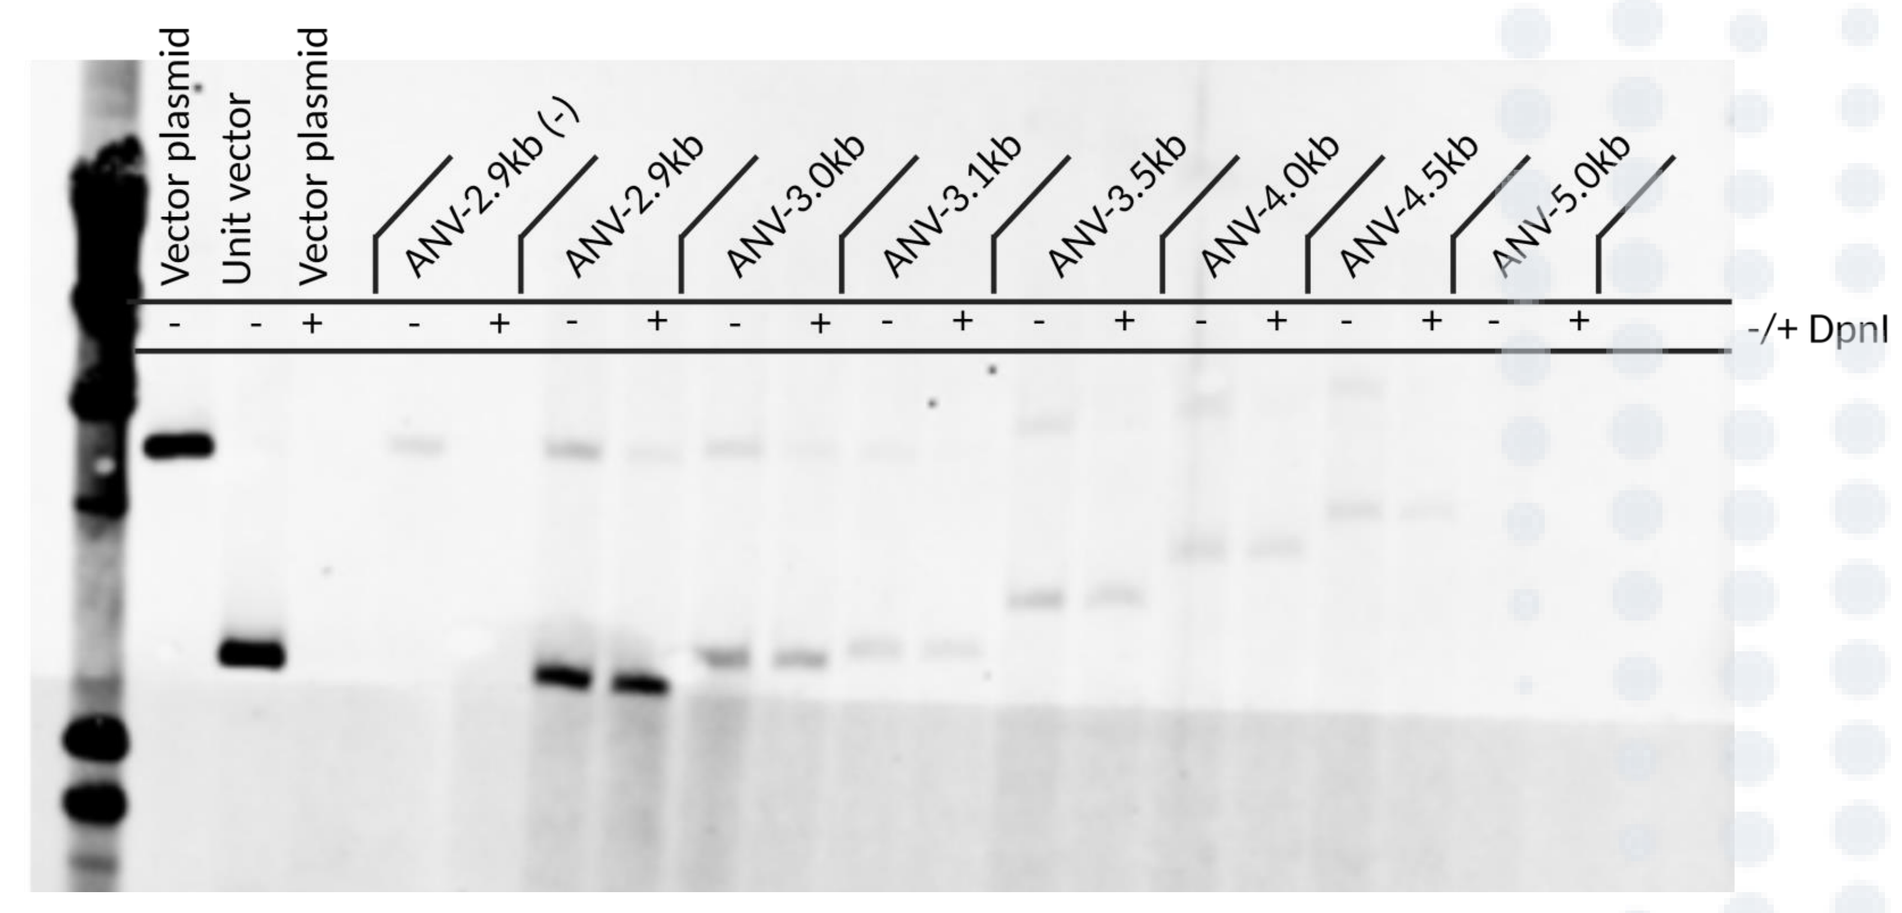
Vector plasmid
Unit vector
Vector plasmid
ANV-2.9kb (-)
ANV-2.9kb
ANV-3.0kb
ANV-3.1kb
ANV-3.5kb
ANV-4.0kb
ANV-4.5kb
ANV-5.0kb
-/+ DpnI

C

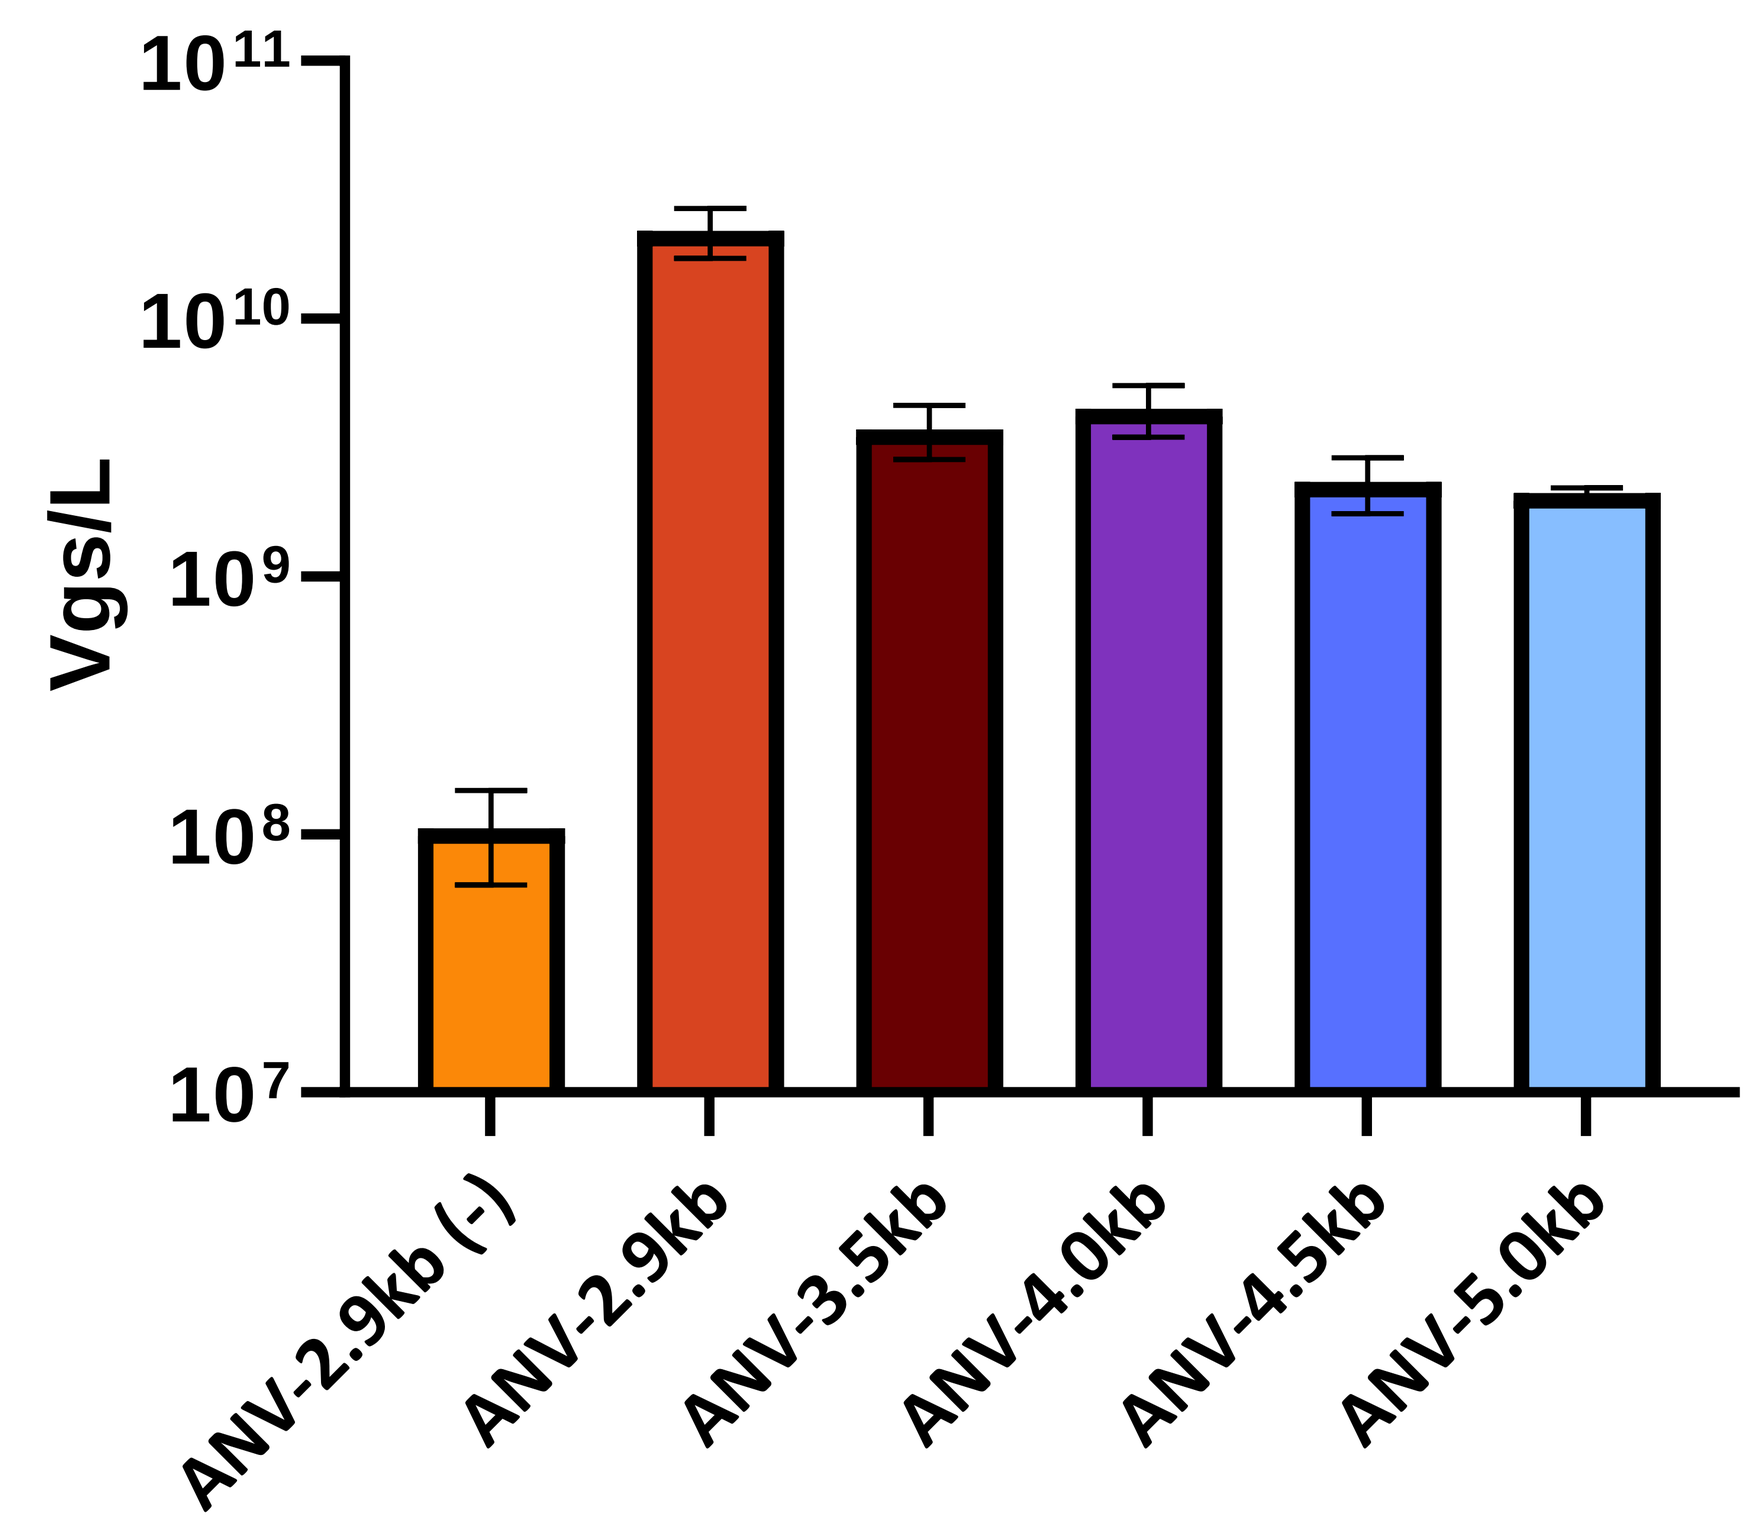
Vgs/L
10^{11}
10^{10}
10^{9}
10^{8}
10^{7}
ANV-2.9kb (-)
ANV-2.9kb
ANV-3.5kb
ANV-4.0kb
ANV-4.5kb
ANV-5.0kb

Figure 3: *AnelloVector™* viral vectors can replicate genomes larger than wild type and package them into viral-like particles A) Schematic representation of expanding the size of a payload in an *AnelloVector™* viral vector by altering the size of random stuffer DNA while keeping the viral non-coding region and eGFP expression transgene constant. This method replicates larger vector genomes and packages into similarly-sized capsid particles using the same viral proteins across conditions. B) Southern blot probing for the eGFP gene in the vector; (+) symbol indicates the sample was digested with DpnI, an enzyme which cuts at its recognition site only where the DNA is bacterially methylated, to remove input plasmid. C) Vectors with payloads up to 5.0kb in size were recovered using isopycnic centrifugation and titer was measured for DNase-protected vector DNA. n = 3

A

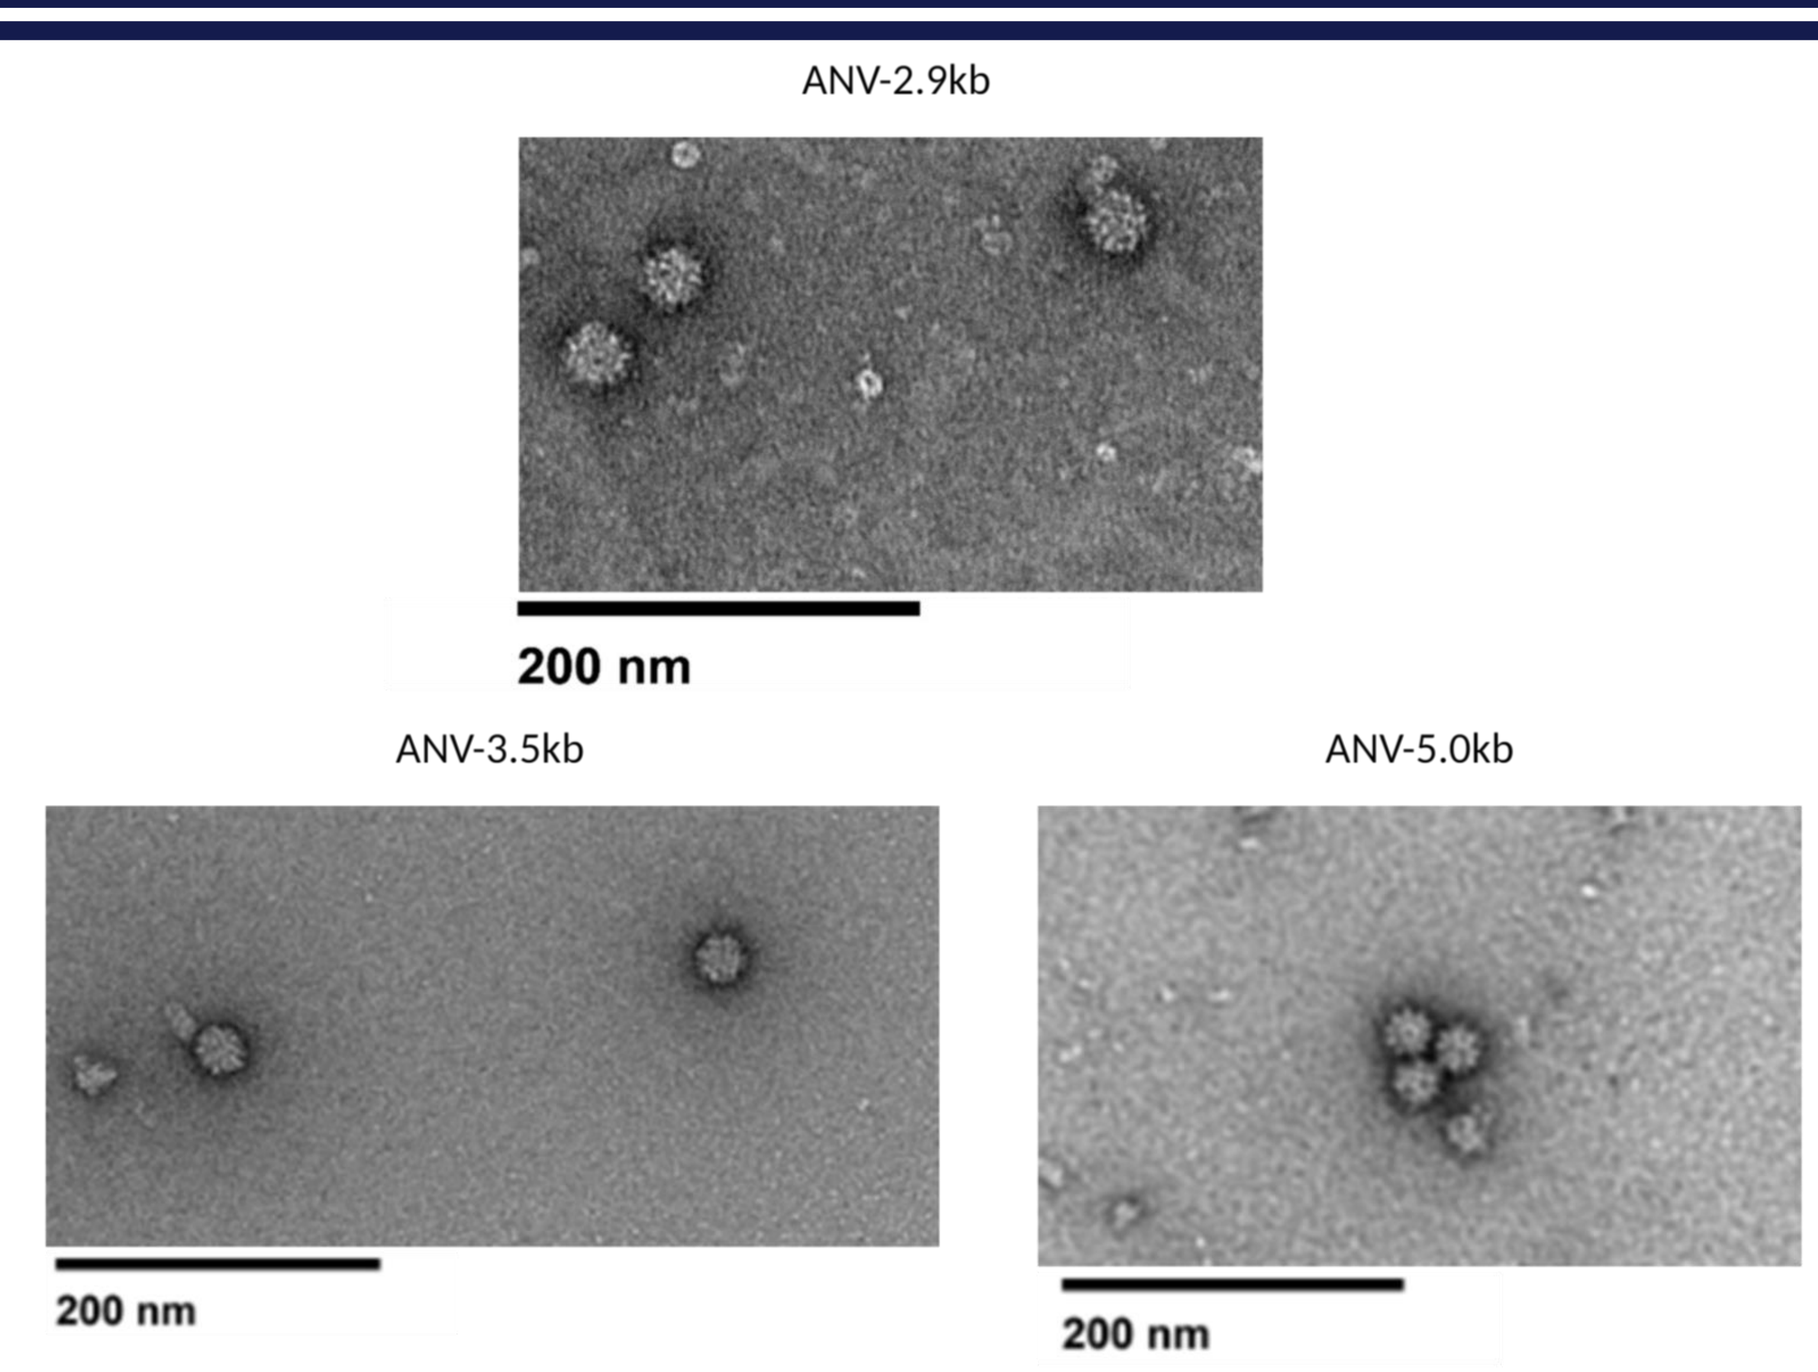
ANV-2.9kb
200 nm
ANV-3.5kb
200 nm
ANV-5.0kb
200 nm

B

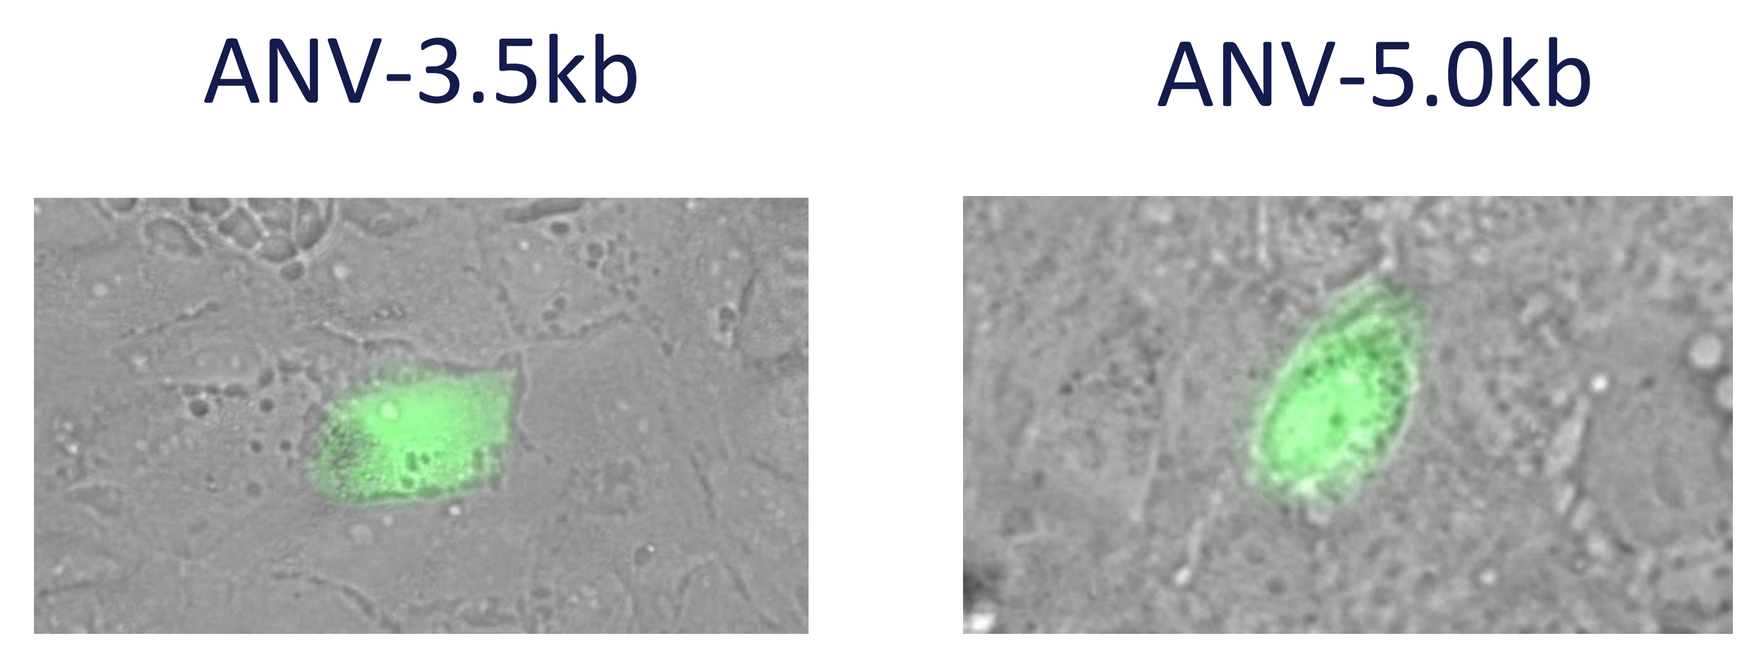
ANV-3.5kb
ANV-5.0kb

Figure 4: Verification of VLPs electron microscopy and transduction. A) Electron micrograph of VLPs containing payloads ranging from 2.9kb to 5.0kb showing particles 30nm in size. Scale bar = 200nm. B) ANV-3.5kb and ANV-5.0kb were transduced in RPE at MOI 2.8K vg/cell and MOI 5.1K, respectively. Cells were imaged on day 7.

AnelloVector™ viral vectors demonstrate 12-month durable expression in vivo post subretinal administration

A

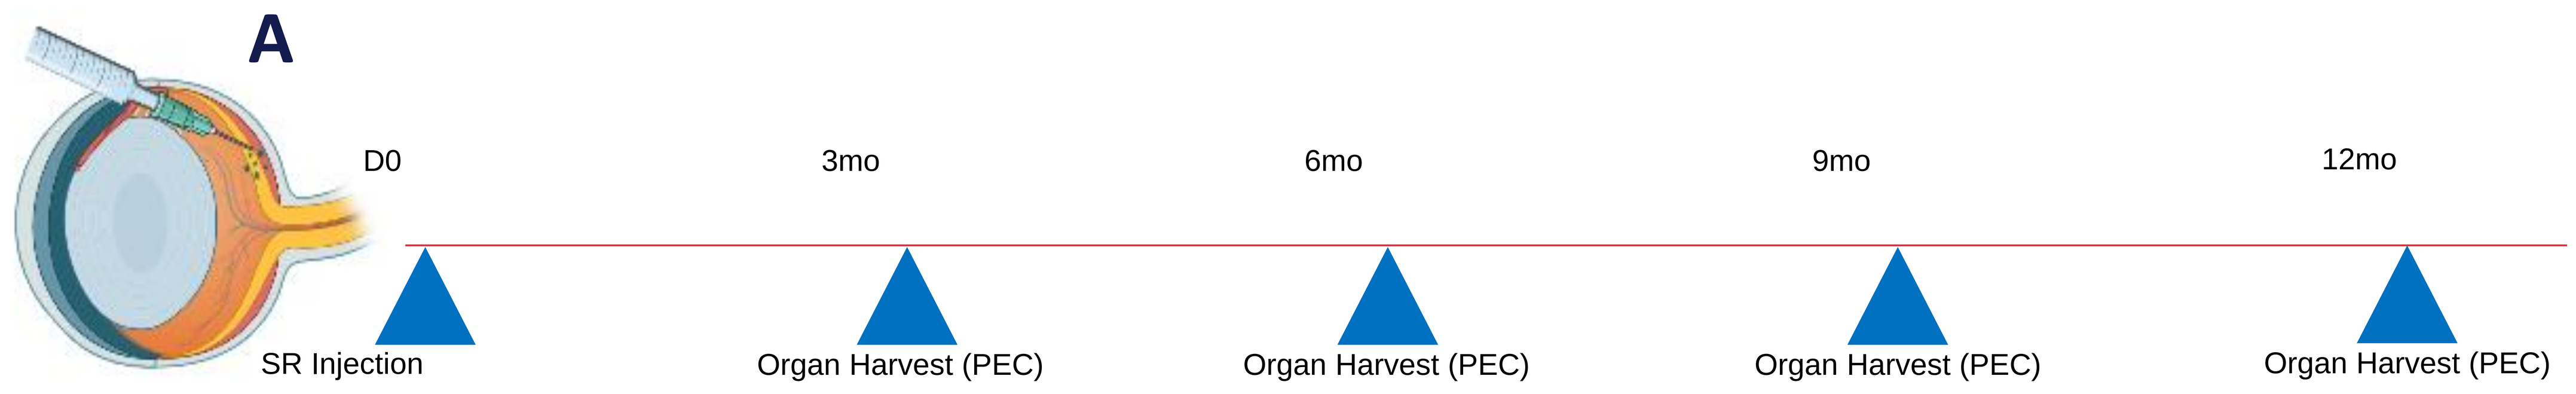
D0
3mo
6mo
9mo
12mo
SR Injection
Organ Harvest (PEC)
Organ Harvest (PEC)
Organ Harvest (PEC)
Organ Harvest (PEC)

B

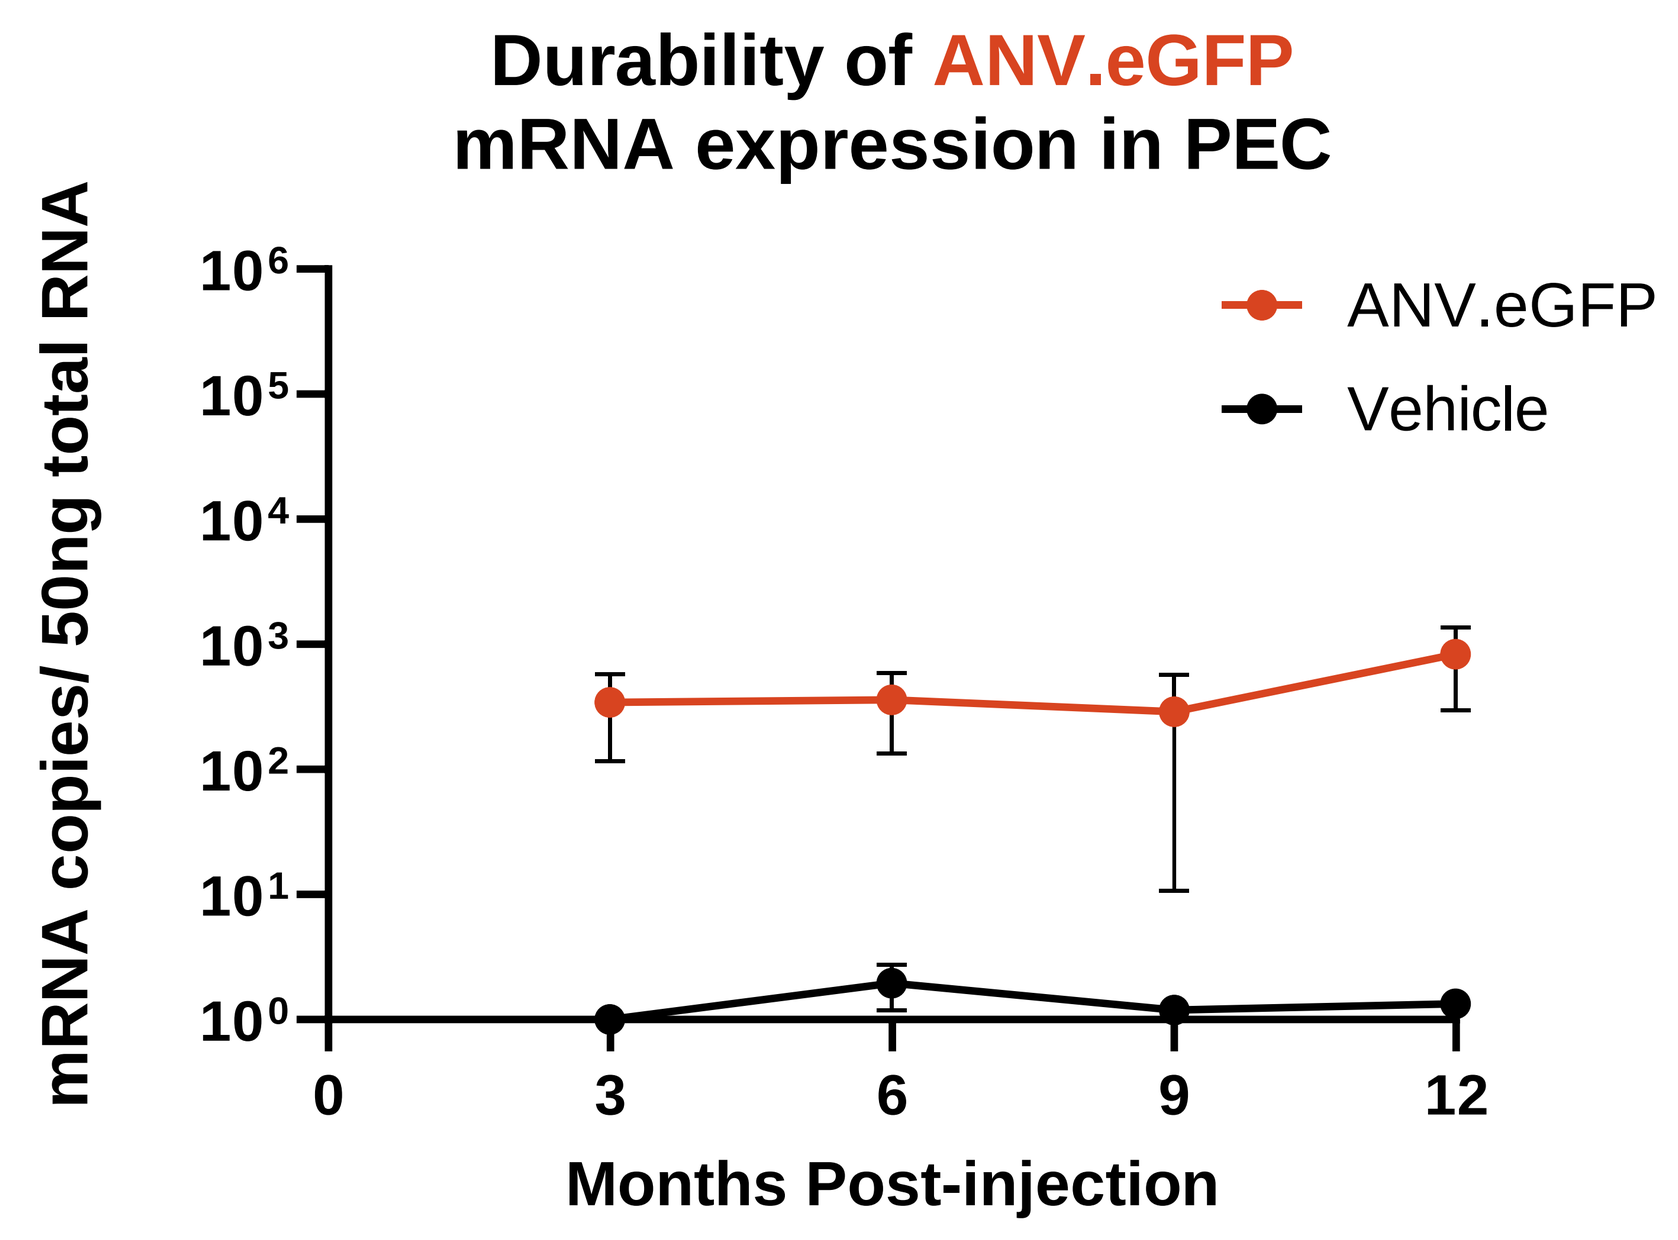
Durability of ANV.eGFP mRNA expression in PEC
mRNA copies/ 50ng total RNA
10^{6}
10^{5}
10^{4}
10^{3}
10^{2}
10^{1}
10^{0}
0
3
6
9
12
Months Post-injection
ANV.eGFP
Vehicle

C

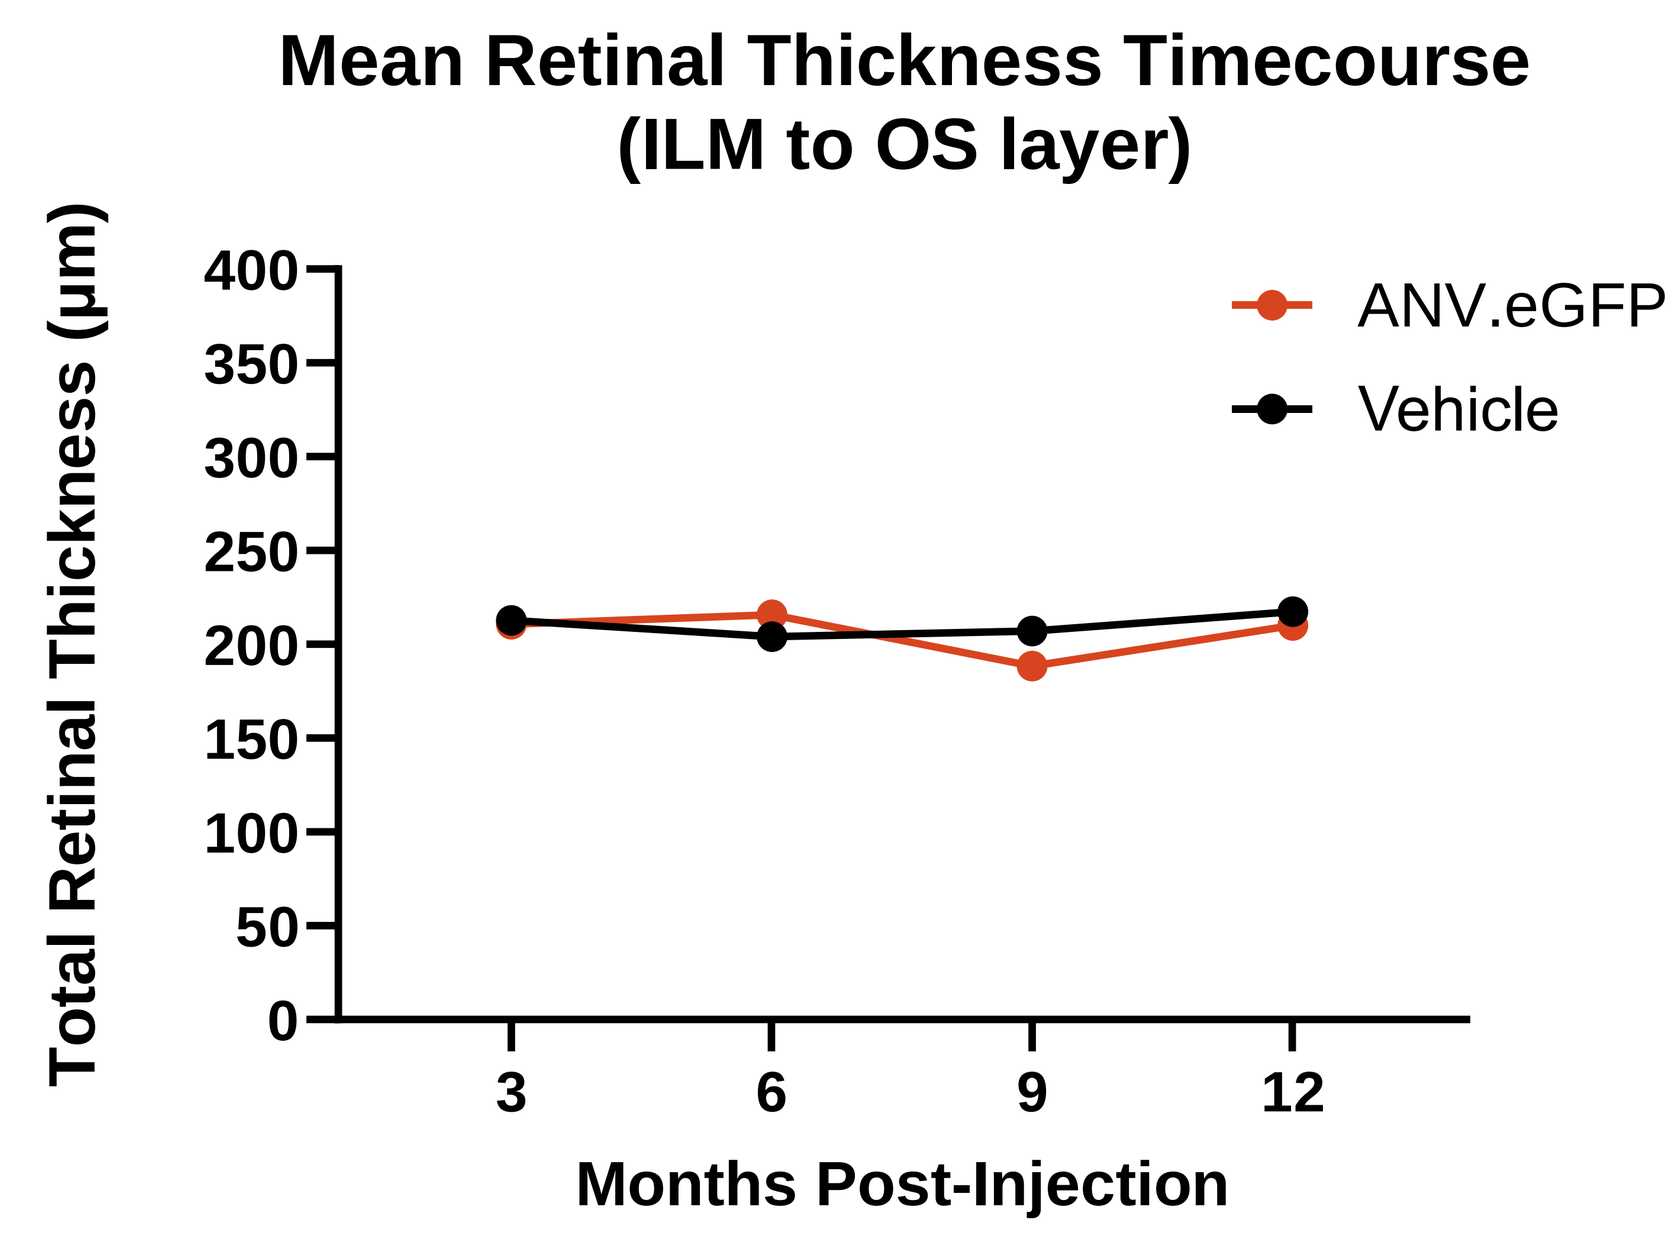
Mean Retinal Thickness Timecourse (ILM to OS layer)
Total Retinal Thickness (µm)
400
350
300
250
200
150
100
50
0
3
6
9
12
Months Post-Injection
ANV.eGFP
Vehicle

D

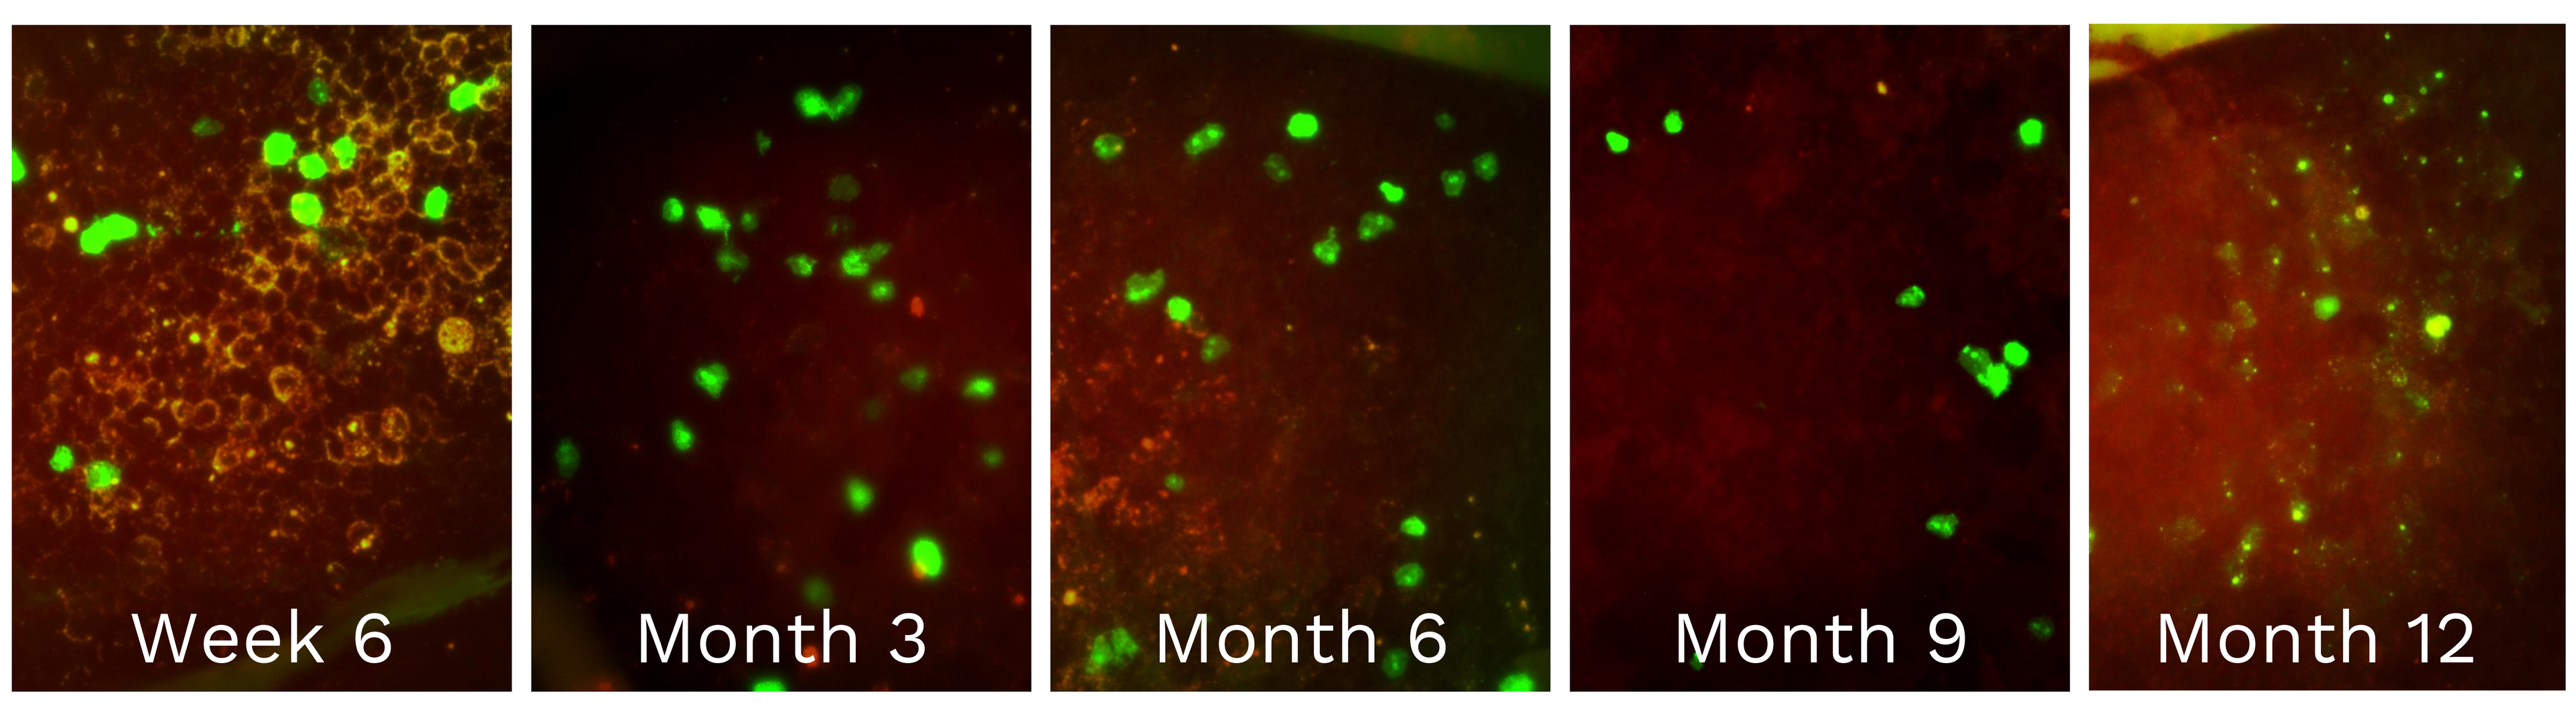
Week 6
Month 3
Month 6
Month 9
Month 12

Figure 5: *AnelloVector™* viral vectors demonstrate durable expression in vivo. A) schematic of a mouse eye's anatomy and study design. B) Absolute mRNA copies present in PEC, as assessed by RT-ddPCR in the eyes injected subretinally with either vehicle or 1E+8 vg of *AnelloVector™* viral vector. n = 4 eyes/group. Error bars indicate SEM. C) Mean retinal thickness measurements of OCT images from C57Bl/6J female mice at 3-, 6- and 9-months post-subretinal injection of either ANV.eGFP vector or vehicle. Measurements represent the distance between the ILM and the OS layers on each eye (n=5-10 eyes/timepoint) at specified intervals. Error bars indicate SEM. D) Flatmount images of PEC taken at the times indicated.